Comparative evaluation of mAb biosimilar development and licensing regulations in BRICS-TM market

Hasumati Rahalkar[1], [2], Hacer COŞKUN Cetintas[3], Sam Salek[1]

[1]School of Life & Medical Sciences, University of Hertfordshire, Hatfield, UK,
[2]Department of Regulatory Sciences, Metina PharmConsulting Pvt Ltd, Mumbai, Maharashtra, India,
[3]Department of Marketing Authorization of Medicines, Turkish Medicines & Medical Devices Agency, Ankara, Turkey

INTRODUCTION

Biosimilars are biotherapeutic products with identical quality and similar safety and efficacy profiles as the reference biological product. It is essential that the standard of evidence supporting the decisions to grant marketing authorization for biosimilars be sufficient to ensure that the products meet acceptable levels of quality, safety and efficacy for public health purposes.

Although the European Medicines Agency (EMA), the United States Food and Drug Administration (USFDA) and the World Health Organization (WHO) have issued specific guidelines with questions and answer documents clarifying doubts pertaining to development, many other agencies are yet to develop mAb specific regulatory guidance.

AIM

The aim was to evaluate EMA, WHO, USFDA, BGTD/HC mAb biosimilar guidelines for development and licensing and compared with BRICS-TM regulations.

METHODS

The current and valid English-language guidelines including published questions and answers such as the EMA guidelines pertaining to biosimilar medicinal product and mAbs, technical report series (TRS) and pertinent annexes of the WHO, specifically for mAbs, guidance for industry from USFDA, the guidance document and the Fact sheet issued by HC/BGTD, MCC (currently known as SAHPRA) /South Africa guidance document and guidelines on similar biologics from India which were obtained from the official websites of the respective regulatory agencies. The authenticated translated guidelines were referred for Brazil, Russia, China, Turkey and Mexico.

RESULTS

Figure 1: Overview of mAb biosimilar development

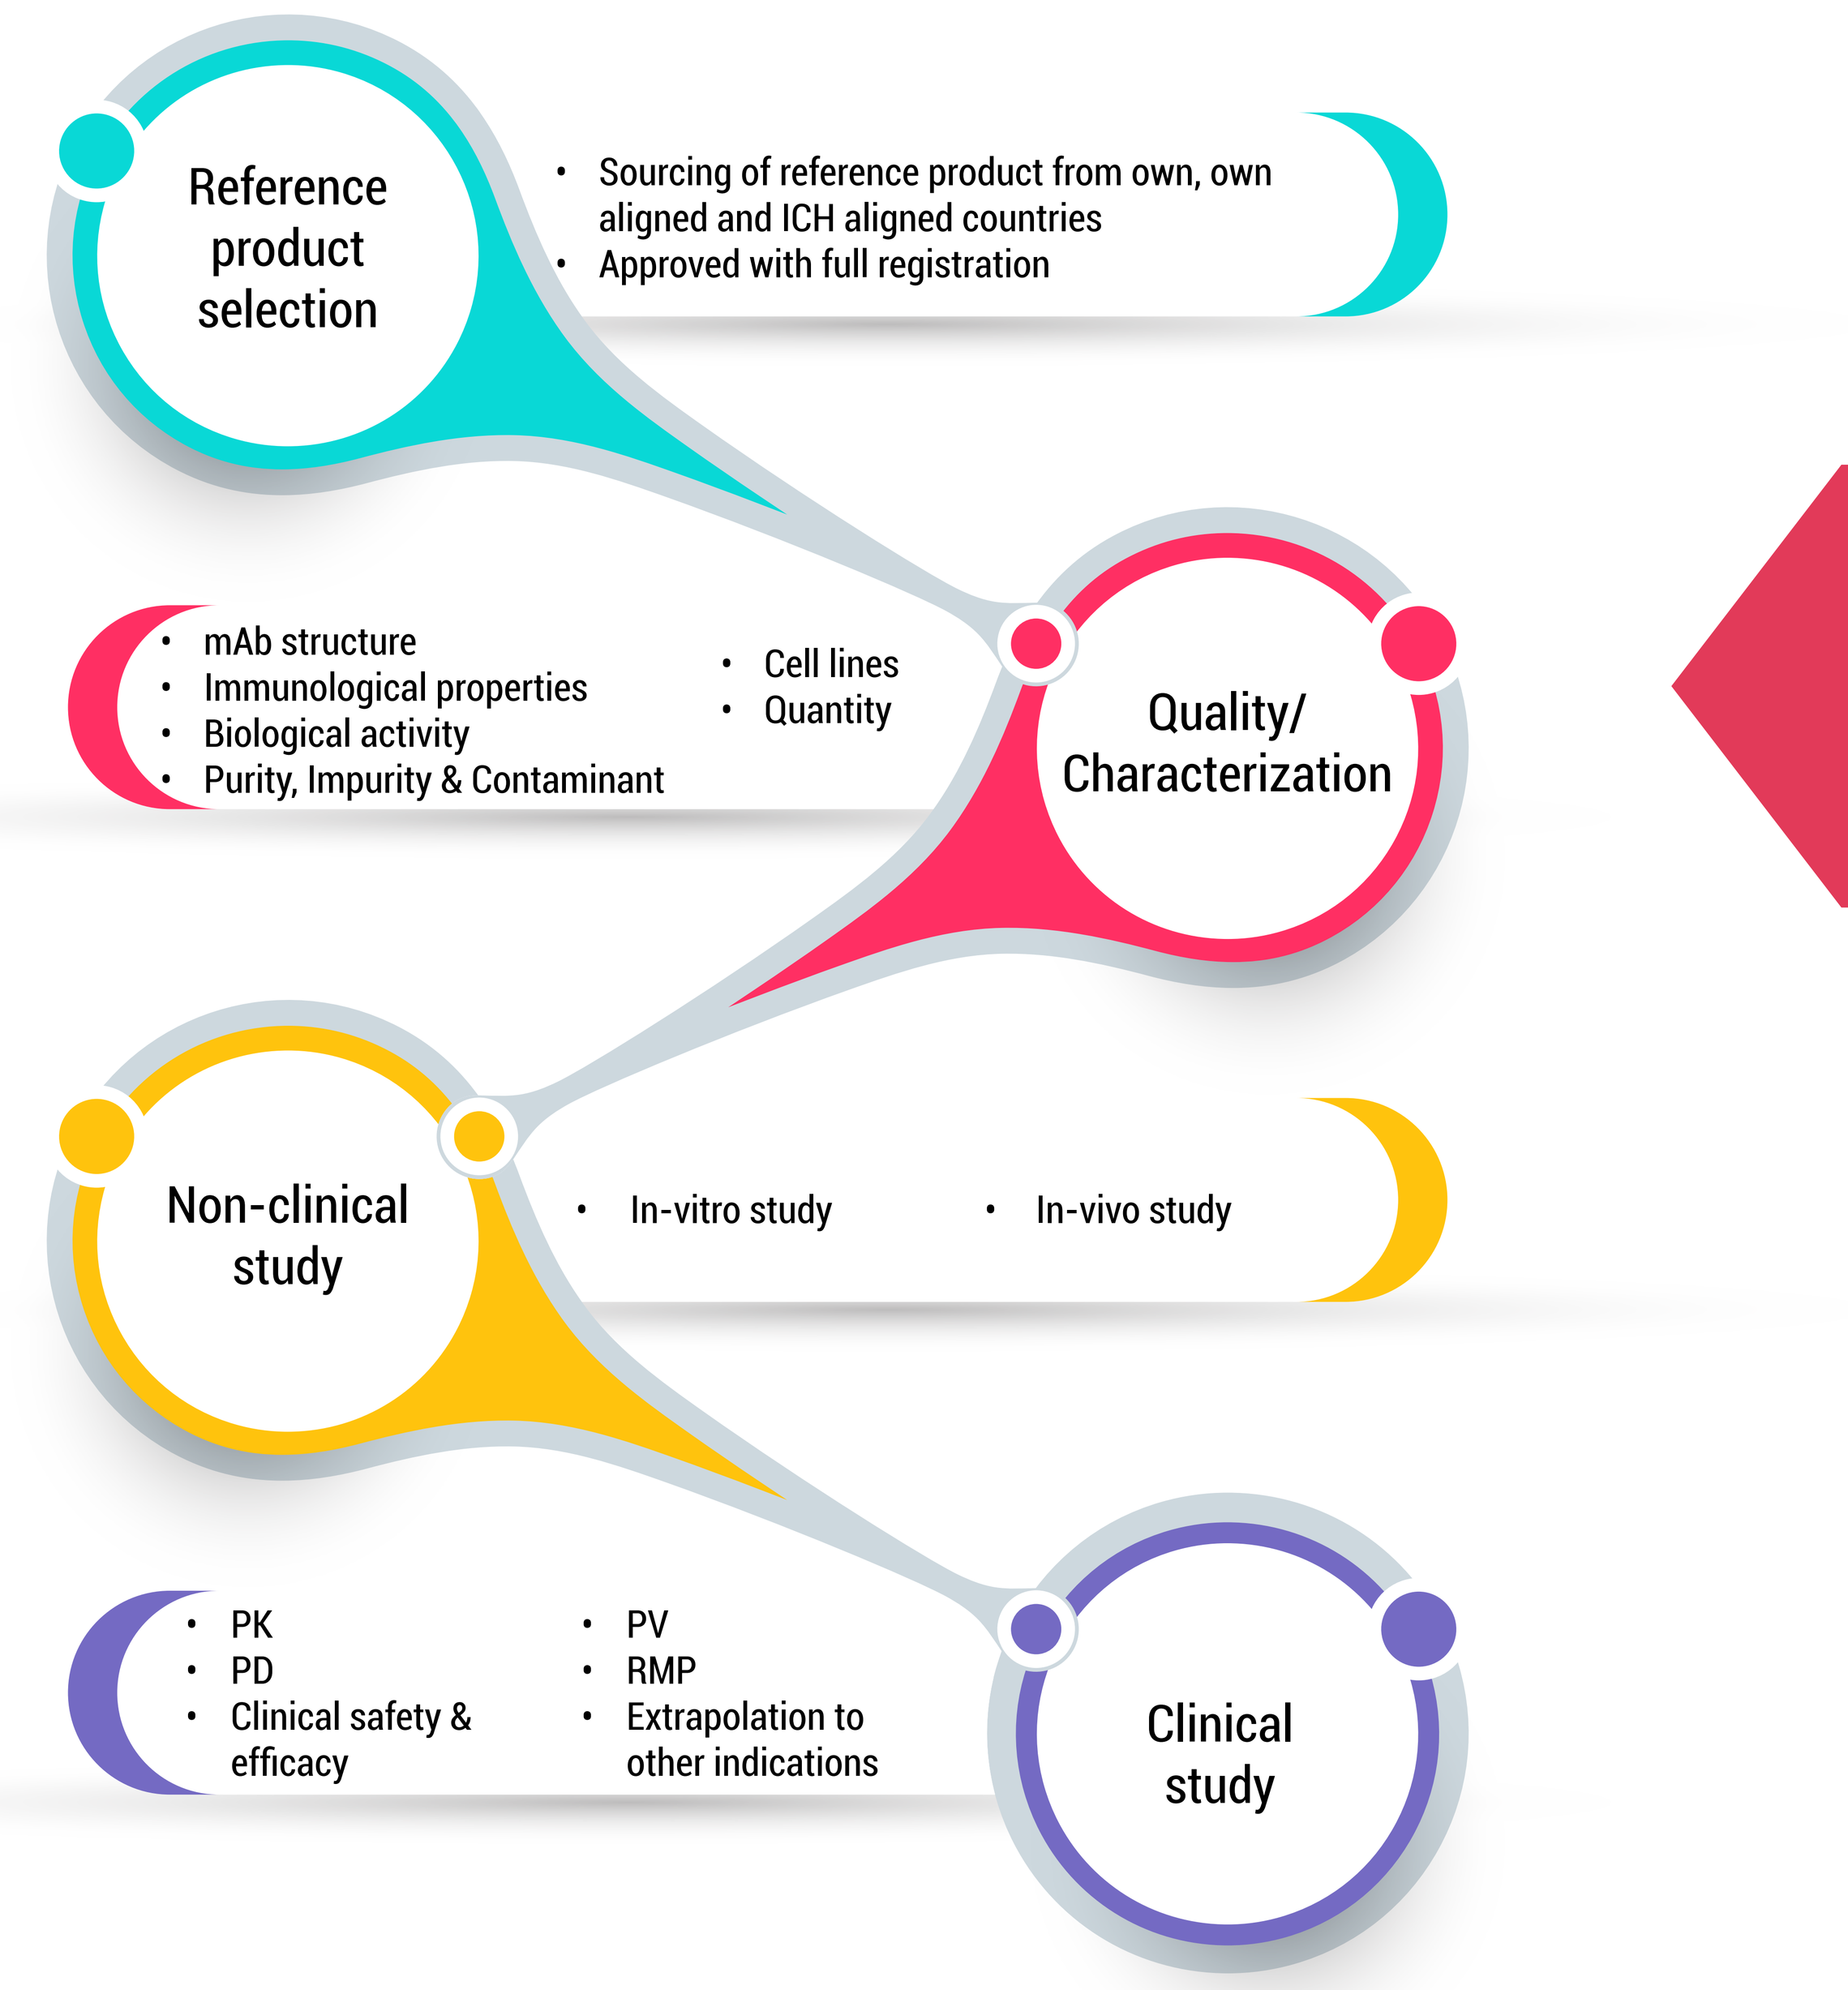
Reference product selection
Sourcing of reference product from own, own aligned and ICH aligned countries
Approved with full registration
mAb structure
Immunological properties
Biological activity
Purity, Impurity & Contaminant
Cell lines
Quantity
Quality/ Characterization
Non-clinical study
In-vitro study
In-vivo study
PK
PD
Clinical safety & efficacy
PV
RMP
Extrapolation to other indications
Clinical study

Figure 2: Publication of mAb specific guidelines

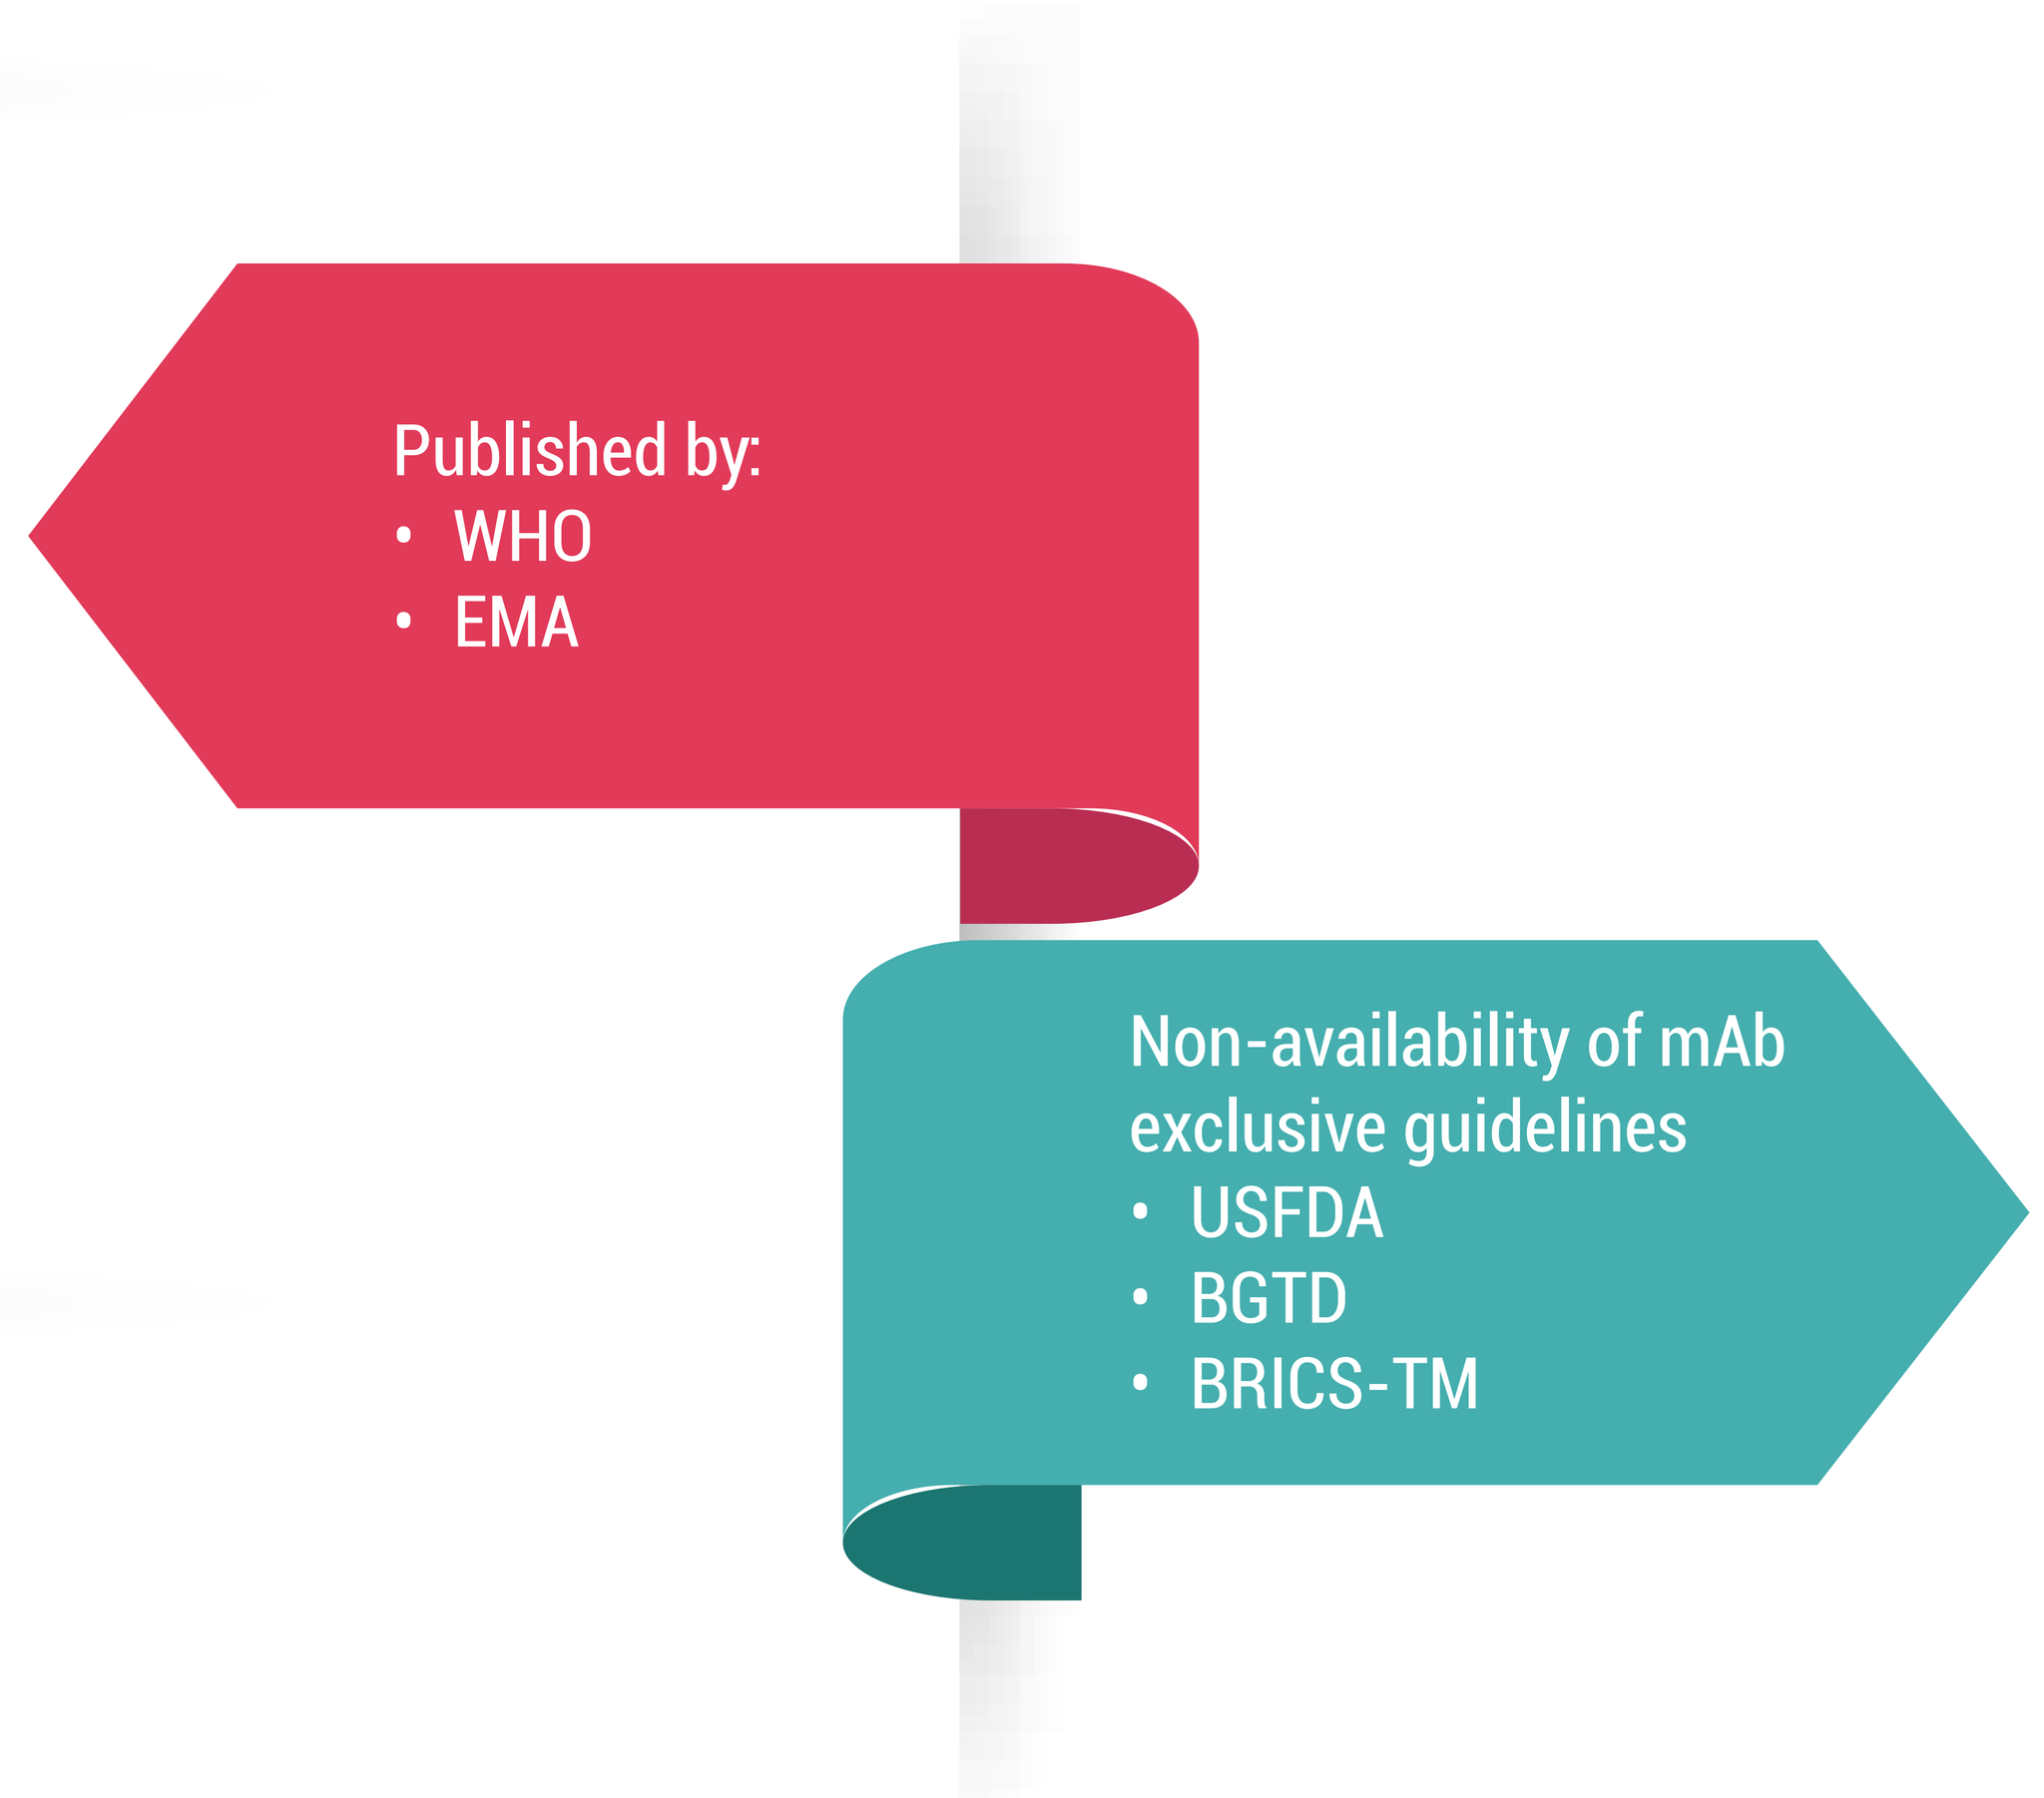
Published by:
WHO
EMA
Non-availability of mAb exclusive guidelines
USFDA
BGTD
BRICS-TM

Table 1: Comparison of reference product selection in well-established agency and BRICS-TM markets.

Agency	Reference product mandates: Licensed in own country	Authorized in ICH countries	Similar or aligned regulatory agency	Bridging data
EMA	✓	✓	×	✓
WHO	×	✓	×	×
USFDA	✓	✓	×	✓
BGTD	✓	✓	✓	×
ANVISA	✓	×	✓	×
Russian federation	✓	×	×	×
CDSCO	✓	✓	×	×
CFDA	✓	×	×	×
SAHPRA	✓	×	✓	×
TMMDA	×	×	✓	×
COFEPRIS	✓	×	×	×

Figure 3: Comparative non-clinical attributes across EMA, WHO, USFDA, BGTD and BRICS-TM markets.

Non clinical	EMA (EU)	WHO	USFDA (USA)	BGTD (Canada)	ANVISA (Brazil)	Russian federation (Russia)	CDSCO (India)	CFDA (China)	SAHPRA (South Africa)	TMMDA (Turkey)	COFEPRIS (Mexico)
In vitro	Comparative binding & functional assays	Comparative binding & functional assays	Functional assay	Performed	n/d	n/d	Cell based assay	n/d	Binding & functional assay	Binding & functional assay	n/d
In vivo											
PKPD	Dose concentration-response assessment	Dose response assessment	Performed	n/d	Mandatory	n/d	n/d	Comparative PKPD	n/d	Dose concentration response assessment	n/d
Toxicity	Repeat dose	Repeat dose	Performed	n/d	Repeat dose	n/d	Repeat dose (1X HED)	Single & Repeat dose	Repeat dose	Repeat dose (flexible approach if non-human primate); Unspecific (for relevant species)	n/d
Immunogenicity	Withdraw blood sample for PK/TK	Withdraw blood sample for PK/TK	Help to interpret animal results	n/d	n/d	n/d	Comparative Ab response	n/d	Comparative bioactivity	Non predictive in human, use for PKTK evaluation	n/d
Safety	n/d	Performed	n/d	n/d	n/d	n/d	n/d	Comparative safety data	Comparative toxicity	n/d	n/d
Local tolerance	for novel excipients	Performed	n/d	n/d	n/d	n/d	Performed	n/d	n/d	For novel excipients	n/d

n/d: Not define, HED: Human equivalent dose

Table 2: Differences in biosimilarity principles across well–established agency and BRICS-TM markets.

Biosimilarity Criteria	EMA(EU)	WHO	USFDA (USA)	BGTD (Canada)	ANVISA (Brazil)	Russian federation (Russia)	CDSCO (India)	CFDA (China)	SAHPRA (South Africa)	TMMDA (Turkey)	COFEPRIS (Mexico)
Posology	✓	×	✓	n/d	n/d	n/d	✓	n/d	n/d	✓	n/d
ROA	✓	✓	✓	✓	n/d	n/d	✓	n/d	n/d	✓	n/d
Strength, form, formulation	✓	✓	✓	✓*	n/d	n/d	✓$^{\delta}$	n/d	n/d	✓	n/d
Improved efficacy	NS	NS	n/d	n/d	n/d	n/d	n/d	n/d	n/d	NS	n/d
Improved safety	✓	✓	n/d	✓	n/d	n/d	n/d	n/d	n/d	✓	n/d
Extrapolation of indications	✓	✓	✓	✓	✓	n/d	✓	✓	✓	✓	n/d
Biosimilarity post approval	NR	n/d	n/d	n/d	n/d	n/d	n/d	n/d	n/d	NR	n/d
Interchangeability, switching, substitution	✓$^{\alpha}$	✓	✓	✓$^{\beta}$	n/d	n/d	n/d	n/d	✓$^{\varepsilon}$	✓	n/d
Pediatric research	✓	n/d	✓	n/d	n/d	n/d	n/d	n/d	n/d	n/d	n/d

*✓ Indicates the requirements are define in the guidelines, NR: Not required, NS: Not suitable, n/d: Not define, *except formulation, αDecision is in hands with member state, βDecision is in hands with province/territory, δPharmaceutical form and formulation not define, εSubstitution is not allowed and interchangeability is possible with practitioner's decision.*

Table 3: Comparative clinical attributes across EMA, WHO, USFDA, BGTD and BRICS-TM markets.

Clinical	EMA(EU)	WHO	USFDA (USA)	BGTD (Canada)	ANVISA (Brazil)	Russian federation (Russia)	CDSCO (India)	CFDA (China)	SAHPRA (South Africa)	TMMDA (Turkey)	COFEPRIS (Mexico)
Pharmacokinetics											
Lowest therapeutic dose	✓	✓*	✓	✓	×	×	✓*	×	✓	✓	×
Subcutaneous route	✓	×	✓$^{\varepsilon}$	×	×	×	✓	×	✓	✓	×
Sampling (Single dose: first & last; Multiple dose: first dose & steady state)	✓$^{\alpha}$	✓#	✓$^{\alpha}$	×	×	×	✓#	×	✓$^{\alpha}$	✓$^{\alpha}$	×
Design (Single dose cross over; Parallel group)	✓$^{\gamma,\rho}$	✓*	✓$^{\eta\rho}$	✓	×	×	✓$^{\gamma,\rho}$	✓$^{\delta}$	✓$^{\gamma,\rho}$	✓$^{\gamma,\rho}$	×
Primary parameter (Single dose: $AUC_{(0\text{-}inf)}$ Multiple dose: C_{max} & C_{trough})	✓$^{\pi}$	×	×	×	×	×	×	×	✓	✓	×
Secondary parameter (Single dose: C_{max}, T_{max}, Vss, $t_{1/2}$, Multiple dose ; $AUC_{(0\text{-}t)}$, steady state AUC)	✓	×	×	×	×	×	×	×	✓$^{\mu}$	✓$^{\mu}$	×
Acceptable range (%)	80-125	80-125	80-125	90-125$^{\omega}$	×	×	✓	×	✓	✓	×
Pharmacodynamics											
PD marker for combined PKPD	✓	✓	✓£	✓$^{\theta}$	✓	×	✓	✓	✓	×	×
Fingerprinting approach (non-surrogate markers)	✓	×	×	Unclear	×	×	×	×	×	×	×
Clinical efficacy											
Study type (Parallel, random, blinded)	✓€	✓€	✓$^{\Psi}$	✓$^{\Omega}$	✓	×	✓	✓€	×	×	×
Patient population for approved indication	✓	✓	×	×	×	×	×	✓	×	×	×
Equivalence design	✓	✓	×	✓	×	×	✓	✓	✓	×	×
Endpoints	✓	✓	×	×	×	×	×	✓	×	×	×
Comparability margin	✓	✓	×	×	×	×	×	✓	×	×	×
Pediatric population	NR	NR	×	×	×	×	×	×	×	×	×
Clinical safety											
Immunogenicity	✓$^{\beta}$	✓$^{\beta}$	✓	×$^{\beta}$	×	×	✓	✓	✓	×	×
Comparative safety data	✓	✓	✓	✓	×	✓	✓	✓	✓	×	×
Follow-up duration (6-12 months)	✓	✓	×	×	×	×	×	×	✓	×	×

*✓ Indicates the requirements are define in the guidelines, × Indicates the requirements are not indicated in the guidelines, NR: Not required, * Higher dose for mAb clearance characteristics, αSteady state sampling for multiple doses, πAdditional truncated AUC for multiple doses, μFor multiple dose: $AUC_{(0\text{-}t)}$, steady state AUC, £ Single or Composite marker, β Double-blind, parallel analysis, duration minimum 4 weeks, €Double-blind, ΩComparative trial, γSingle dose for late elimination, ¥ Parallel group for mAb characteristics, ρParallel group for long half-life, δ Only single & multiple doses, η Cross over for short half life, ε Same as reference product, θStudy PK/PD relationship, ΨOnly parallel design, #Single dose: Till last quantifiable concentration, ωequivalence margine for primary parameters.*

DISCUSSION

It is evident that mAb biosimilar exclusive guidelines are yet to be published by BRICS-TM agencies. With reference to usage of reference biological medicinal product from ICH or similarly aligned regulatory agencies, the expectations of bridging data is unclear for each of the BRICS-TM market. The necessity of proving biosimilarity post authorization, interchangeability, switching, substitution and pediatric research remains undefined with these emerging agencies. Further non-clinical and clinical mandates with detailed requirements are yet awaited. Though there are gaps in mAb biosimilar regulatory guidelines in emerging markets, we believe that the agencies are working hard to align regulatory norms in line with well-established agencies. The regulator participates in multiple forums, exchanges knowledge and is willing to upgrade. It would be advisable for the companies to approach agencies in advance to obtain biosimilar development advice so that hassle free authorization can be obtained.

ABBREVIATIONS

ANVISA: The Brazilian Health Surveillance Agency, BGTD: Biologics and Genetic Therapies Directorate, BRICS-TM: Brazil Russia India China South Africa- Turkey Mexico, CDR: Complementary Determining Region, CDSCO: Central Drugs Standard Control Organization, CFDA: China Food and Drug Administration, COFEPRIS: The Federal Commission for the Protection against Sanitary Risk, EMA: European Medicines Agency, ICH: International Conference on Harmonisation, mAb: Monoclonal Antibodies, PD: Pharmacodynamics, PK: Pharmacokinetics, RBP: Reference Biological Product, ROA: Route of Administration, SAHPRA: South African Health Products Regulatory Authority, TK: Toxico-kinetics, TMMDA: Turkish Medicines and Medical Devices Agency, USFDA: United States Food and Drug Administration, WHO: World Health Organisation

ACKNOWLEDGMENT

We highly appreciate and acknowledge 'Agnes V. Klein, MD Sr. Medical Advisor, BGTD/HC' for reviewing and supporting with expert comments for Canadian biosimilar regulations.

DISCLOSURE

Authors of this presentation have nothing to disclose.

CONTACT
HASUMATI RAHALKAR
hasumati@metinapharmconsulting.com
Tel- +91 9820113613

Comparative evaluation of mAb biosimilar development and licensing regulations in BRICS-TM market

Hasumati Rahalkar[1], [2], Hacer COŞKUN Cetintas[3], Sam Salek[1]

[1]School of Life & Medical Sciences, University of Hertfordshire, Hatfield, UK,
[2]Department of Regulatory Sciences, Metina PharmConsulting Pvt Ltd, Mumbai, Maharashtra, India,
[3]Department of Marketing Authorization of Medicines, Turkish Medicines & Medical Devices Agency, Ankara, Turkey

INTRODUCTION

Biosimilars are biotherapeutic products with identical quality and similar safety and efficacy profiles as the reference biological product. It is essential that the standard of evidence supporting the decisions to grant marketing authorization for biosimilars be sufficient to ensure that the products meet acceptable levels of quality, safety and efficacy for public health purposes.

Although the European Medicines Agency (EMA), the United States Food and Drug Administration (USFDA) and the World Health Organization (WHO) have issued specific guidelines with questions and answer documents clarifying doubts pertaining to development, many other agencies are yet to develop mAb specific regulatory guidance.

AIM

The aim was to evaluate EMA, WHO, USFDA, BGTD/HC mAb biosimilar guidelines for development and licensing and compared with BRICS-TM regulations.

METHODS

The current and valid English-language guidelines including published questions and answers such as the EMA guidelines pertaining to biosimilar medicinal product and mAbs, technical report series (TRS) and pertinent annexes of the WHO, specifically for mAbs, guidance for industry from USFDA, the guidance document and the Fact sheet issued by HC/BGTD, MCC (currently known as SAHPRA) /South Africa guidance document and guidelines on similar biologics from India which were obtained from the official websites of the respective regulatory agencies. The authenticated translated guidelines were referred for Brazil, Russia, China, Turkey and Mexico.

RESULTS

Figure 1: Overview of mAb biosimilar development

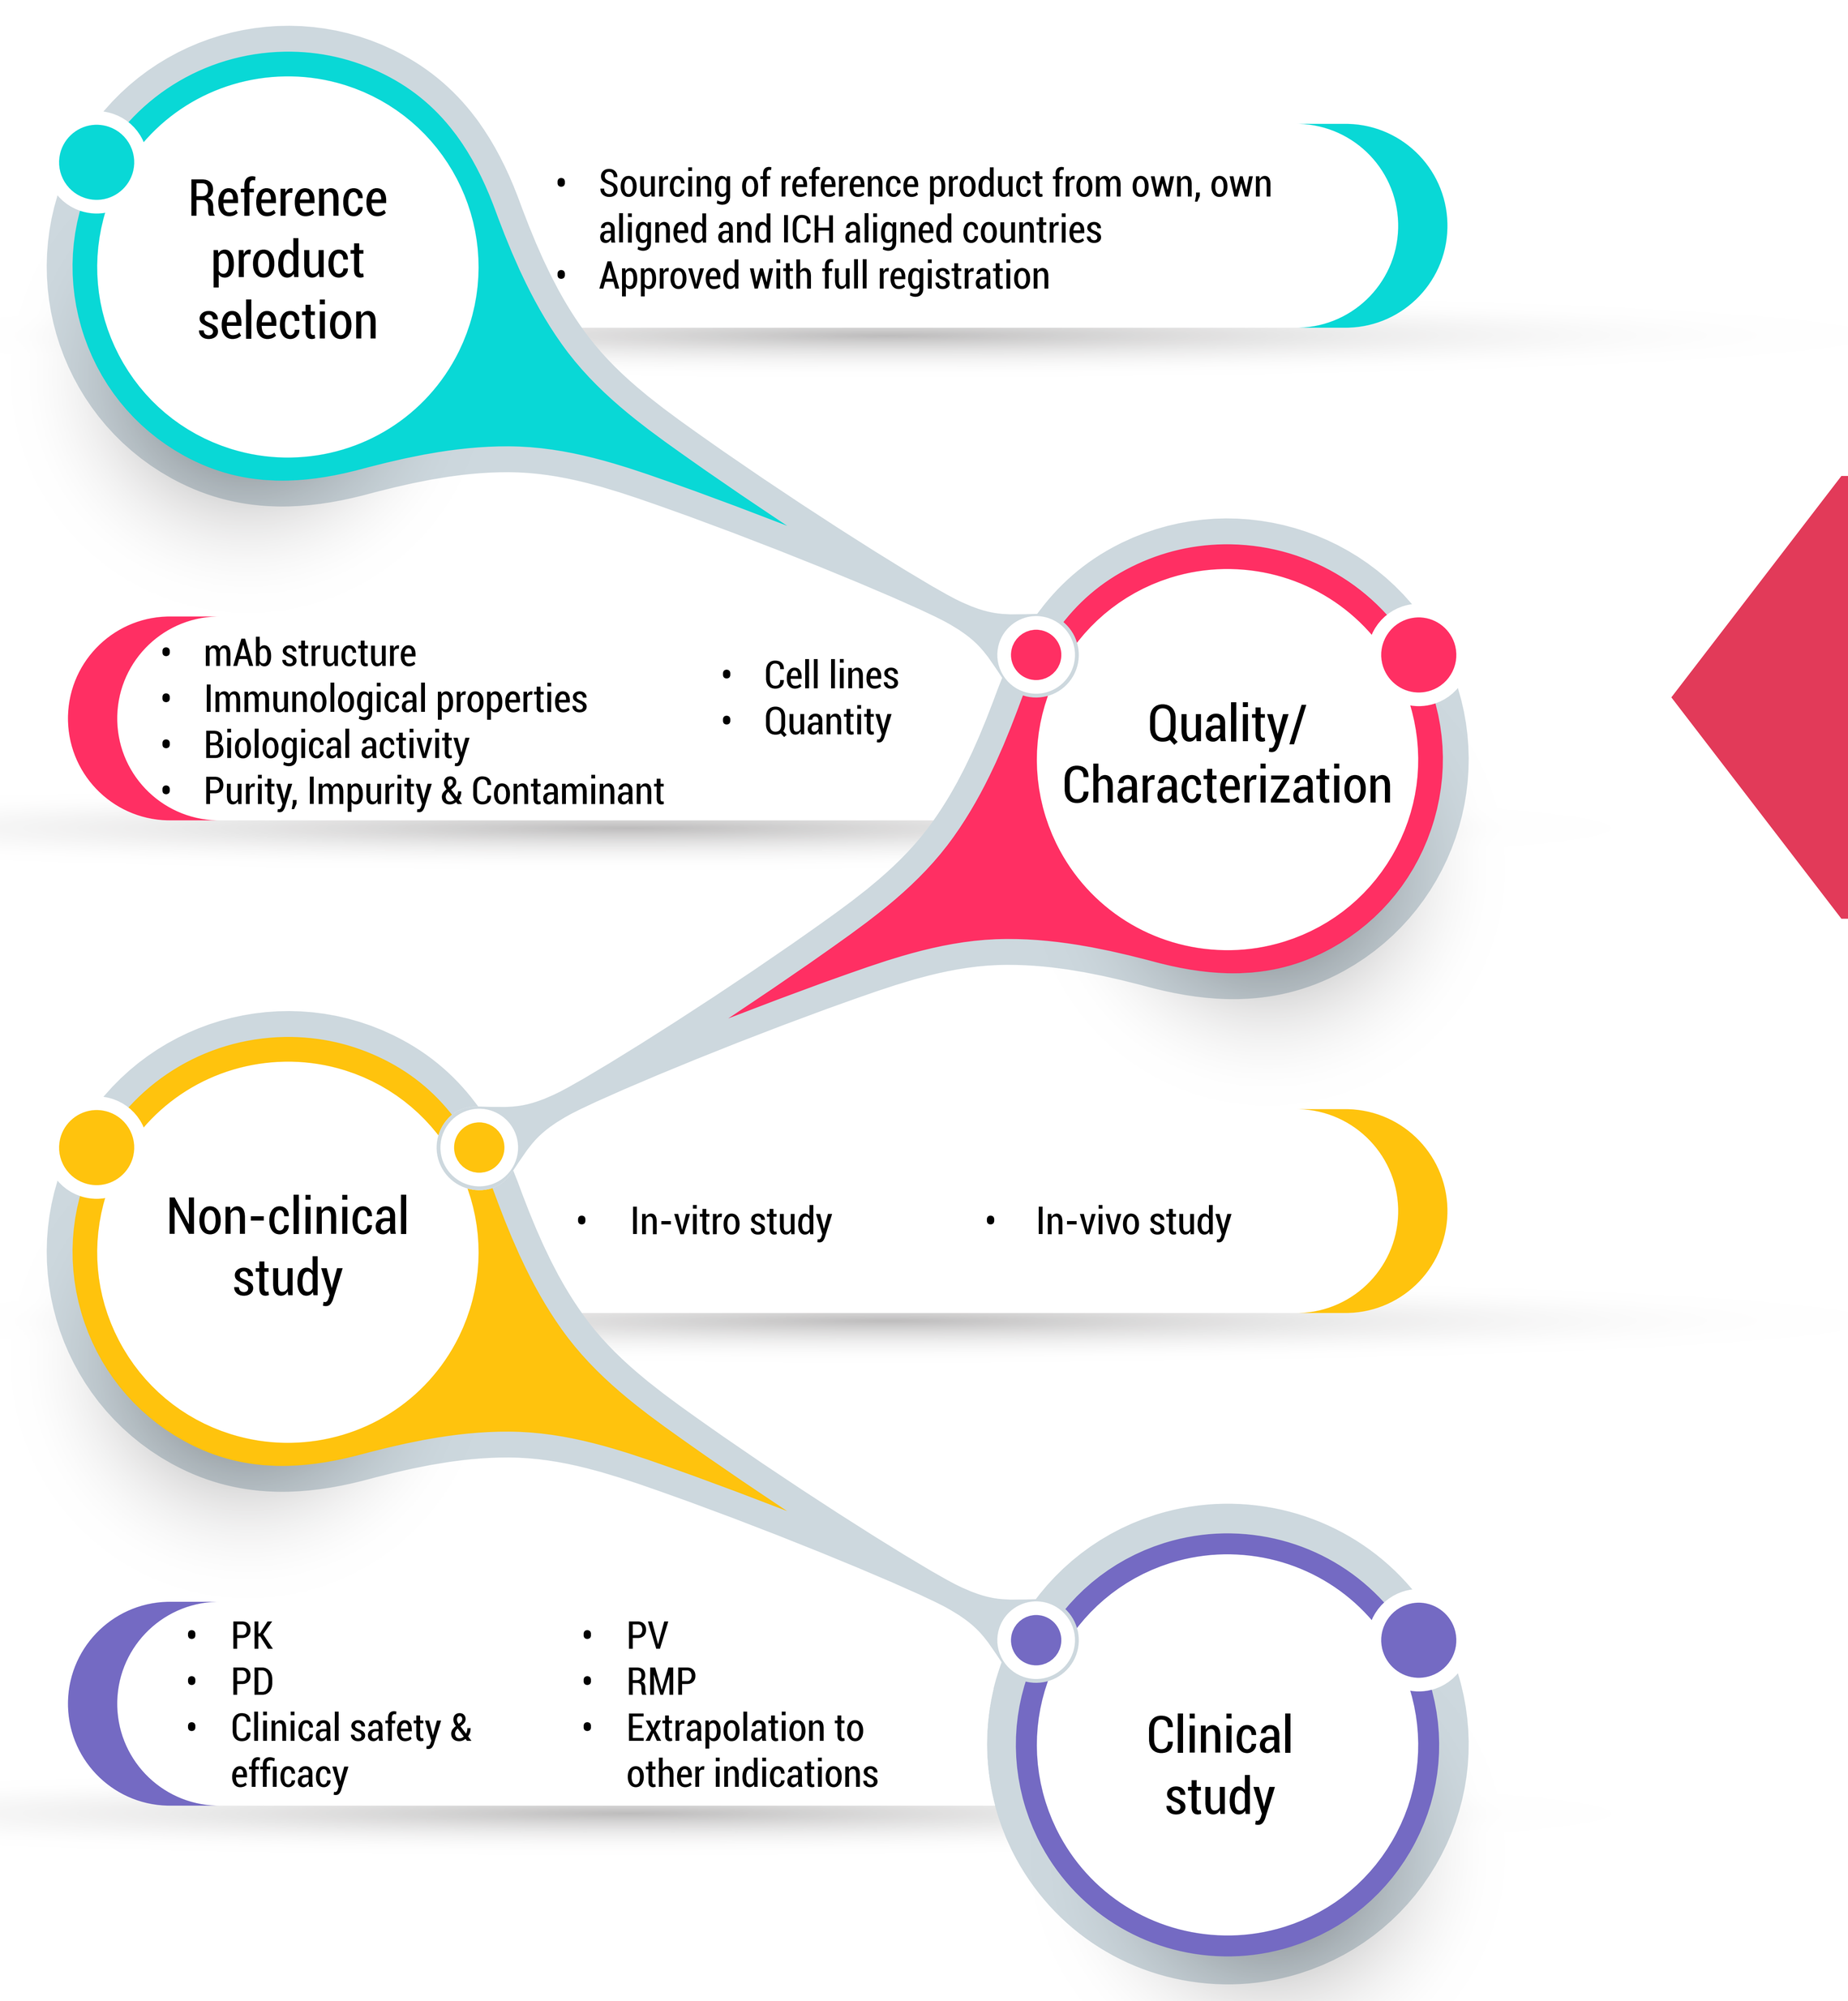

Reference product selection
Sourcing of reference product from own, own aligned and ICH aligned countries
Approved with full registration
mAb structure
Immunological properties
Biological activity
Purity, Impurity & Contaminant
Cell lines
Quantity
Quality/ Characterization
Non-clinical study
In-vitro study
In-vivo study
PK
PD
Clinical safety & efficacy
PV
RMP
Extrapolation to other indications
Clinical study

Figure 2: Publication of mAb specific guidelines

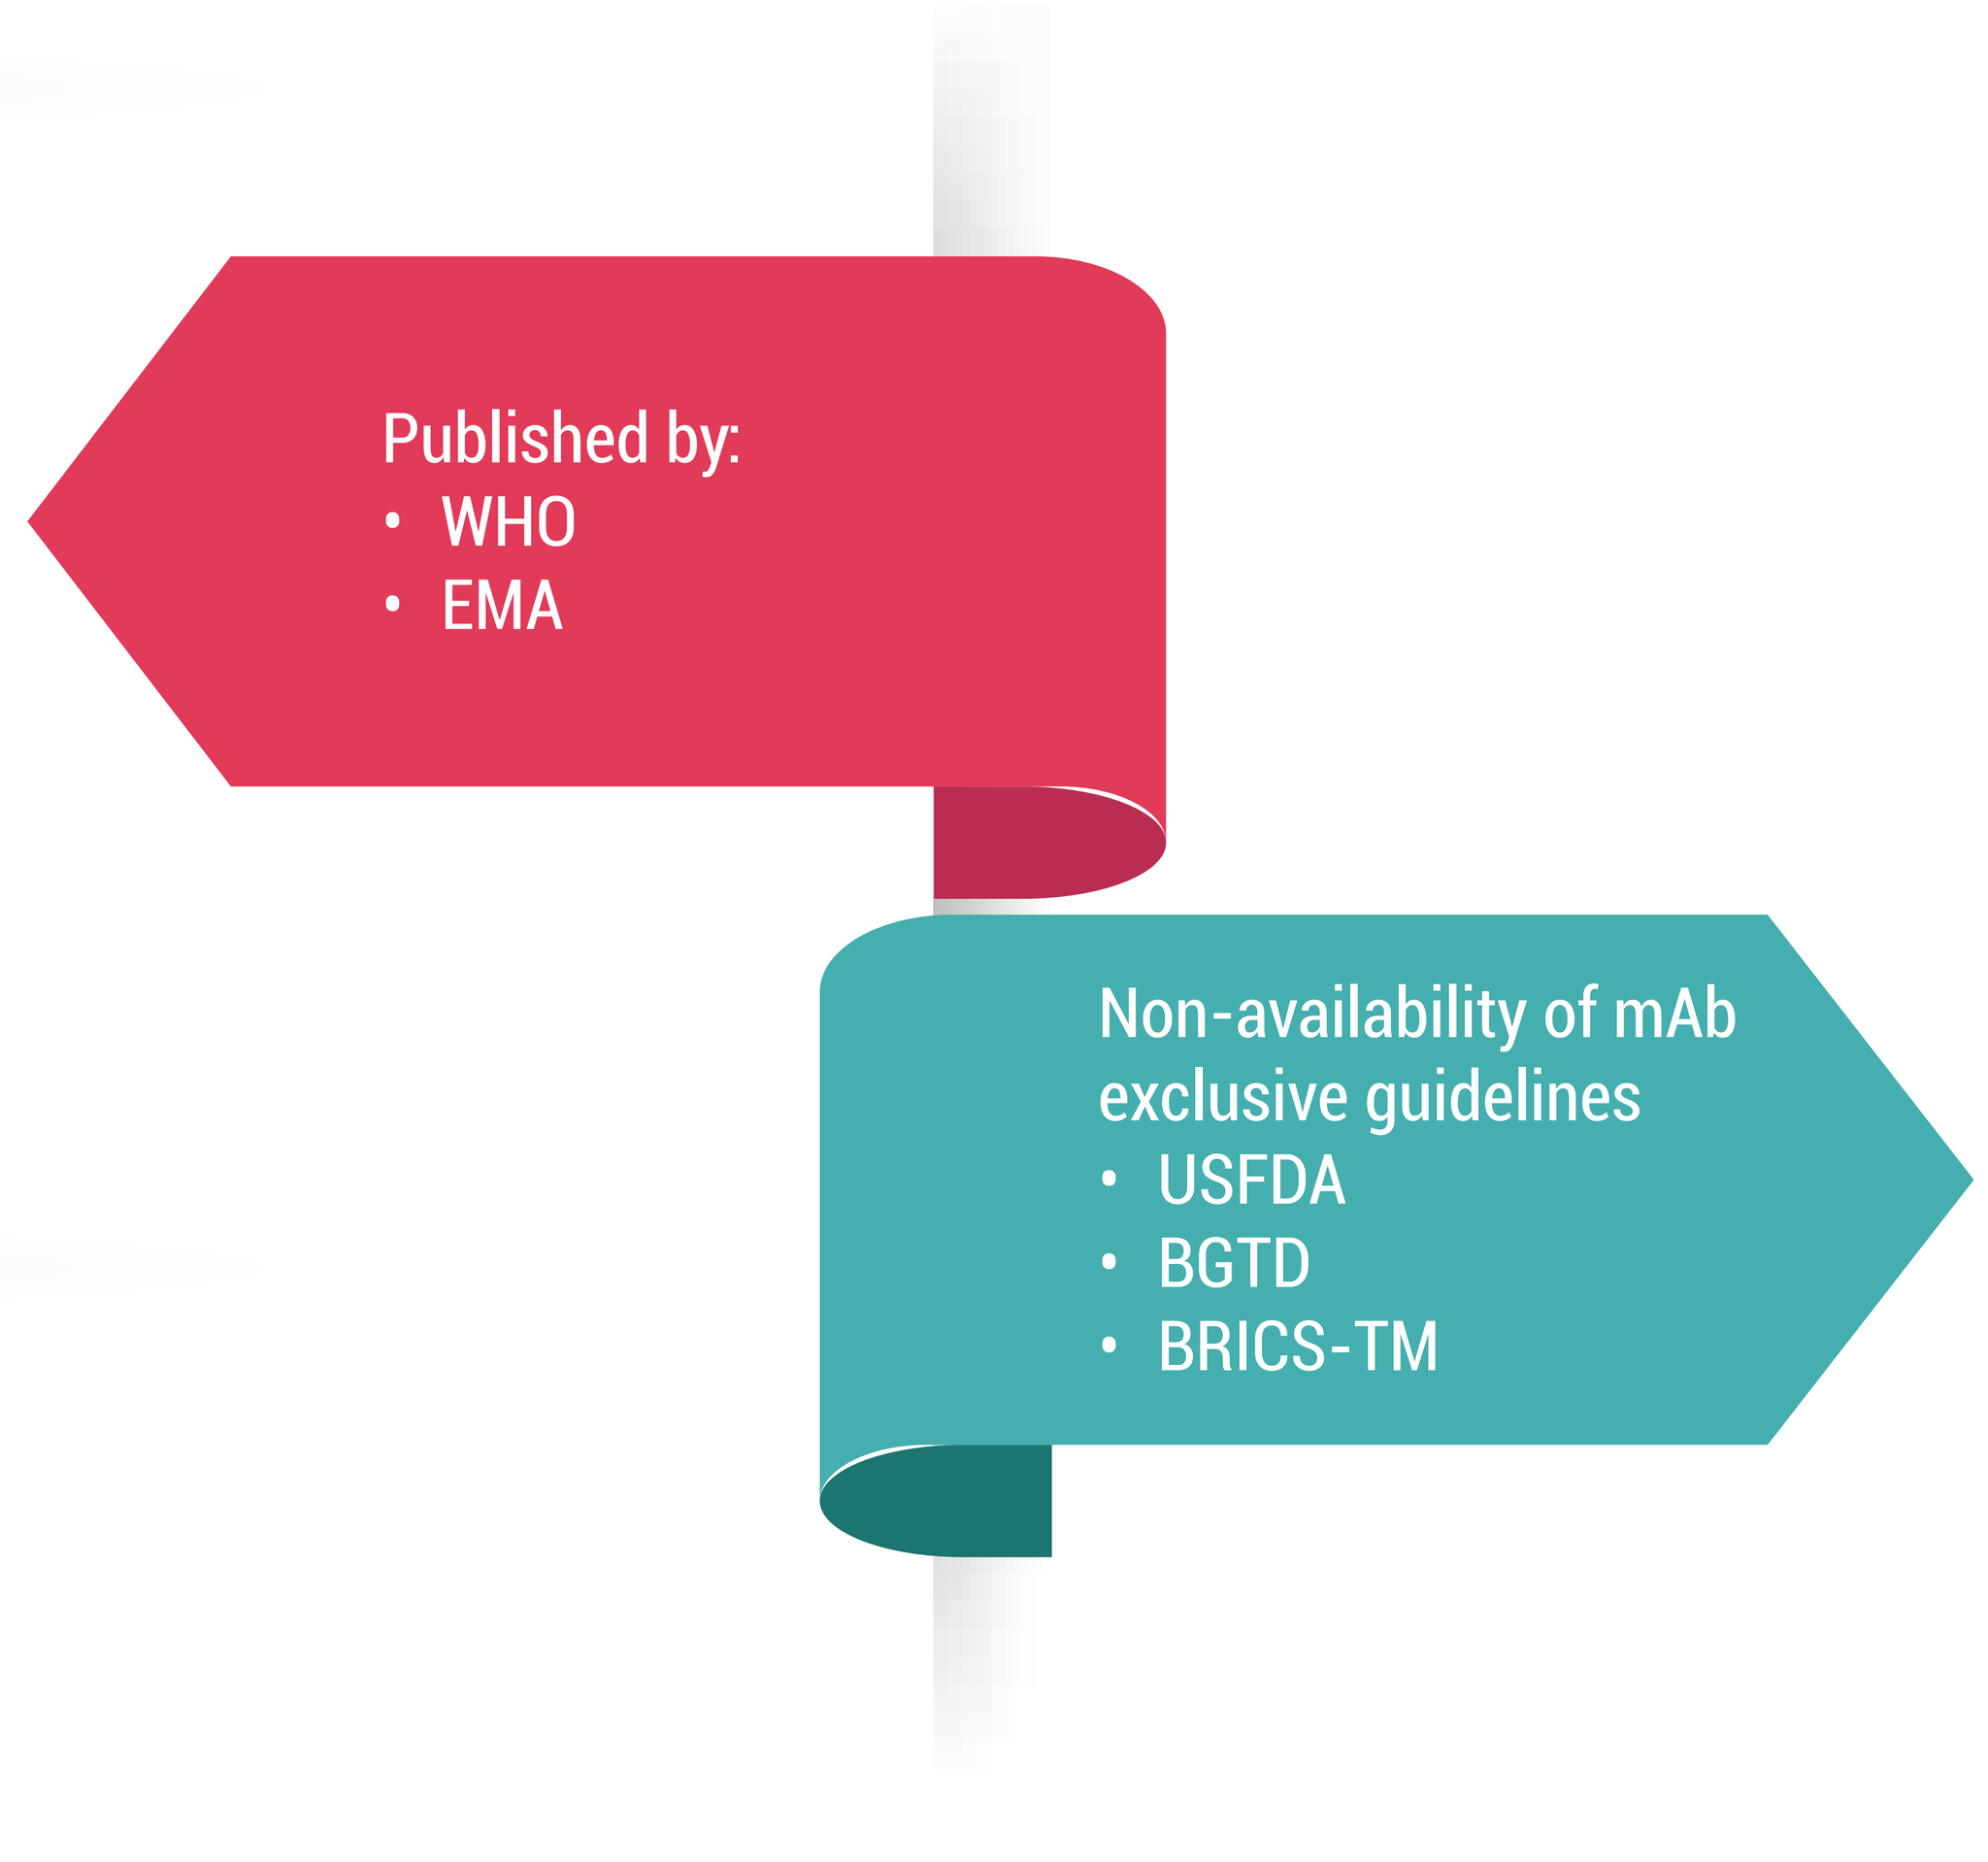

Published by:
WHO
EMA
Non-availability of mAb exclusive guidelines
USFDA
BGTD
BRICS-TM

Table 1: Comparison of reference product selection in well-established agency and BRICS-TM markets.

Agency	Reference product mandates			
	Licensed in own country	Authorized in ICH countries	Similar or aligned regulatory agency	Bridging data
EMA	✓	✓	✗	✓
WHO	✗	✓	✗	✗
USFDA	✓	✓	✗	✓
BGTD	✓	✓	✓	✗
ANVISA	✓	✗	✓	✗
Russian federation	✓	✗	✗	✗
CDSCO	✓	✓	✗	✗
CFDA	✓	✗	✗	✗
SAHPRA	✓	✗	✓	✗
TMMDA	✗	✗	✓	✗
COFEPRIS	✓	✗	✗	✗

Figure 3: Comparative non-clinical attributes across EMA, WHO, USFDA, BGTD and BRICS-TM markets.

Non clinical	EMA (EU)	WHO	USFDA (USA)	BGTD (Canada)	ANVISA (Brazil)	Russian federation (Russia)	CDSCO (India)	CFDA (China)	SAHPRA (South Africa)	TMMDA (Turkey)	COFEPRIS (Mexico)
In vitro	Comparative binding & functional assays	Comparative binding & functional assays	Functional assay	Performed	n/d	n/d	Cell based assay	n/d	Binding & functional assay	Binding & functional assay	n/d
In vivo											
PKPD	Dose concentration-response assessment	Dose response assessment	Performed	n/d	Mandatory	n/d	n/d	Comparative PKPD	n/d	Dose concentration response assessment	n/d
Toxicity	Repeat dose	Repeat dose	Performed	n/d	Repeat dose	n/d	Repeat dose (1X HED)	Single & Repeat dose	Repeat dose	Repeat dose (flexible approach if non-human primate); Unspecific (for relevant species)	n/d
Immunogenicity	Withdraw blood sample for PK/TK	Withdraw blood sample for PK/TK	Help to interpret animal results	n/d	n/d	n/d	Comparative Ab response	n/d	Comparative bioactivity	Non predictive in human, use for PKTK evaluation	n/d
Safety	n/d	Performed	n/d	n/d	n/d	n/d	n/d	Comparative safety data	Comparative toxicity	n/d	n/d
Local tolerance	for novel excipients	Performed	n/d	n/d	n/d	n/d	Performed	n/d	n/d	For novel excipients	n/d

n/d: Not define, HED: Human equivalent dose

Table 2: Differences in biosimilarity principles across well–established agency and BRICS-TM markets.

Biosimilarity Criteria	EMA(EU)	WHO	USFDA (USA)	BGTD (Canada)	ANVISA (Brazil)	Russian federation (Russia)	CDSCO (India)	CFDA (China)	SAHPRA (South Africa)	TMMDA (Turkey)	COFEPRIS (Mexico)
Posology	✓	✗	✓	n/d	n/d	n/d	✓	n/d	n/d	✓	n/d
ROA	✓	✓	✓	✓	n/d	n/d	✓	n/d	n/d	✓	n/d
Strength, form, formulation	✓	✓	✓	✓*	n/d	n/d	✓$^{\delta}$	n/d	n/d	✓	n/d
Improved efficacy	NS	NS	n/d	n/d	n/d	n/d	n/d	n/d	n/d	NS	n/d
Improved safety	✓	✓	n/d	✓	n/d	n/d	n/d	n/d	n/d	✓	n/d
Extrapolation of indications	✓	✓	✓	✓	✓	n/d	✓	✓	✓	✓	n/d
Biosimilarity post approval	NR	n/d	n/d	n/d	n/d	n/d	n/d	n/d	n/d	NR	n/d
Interchangeability, switching, substitution	✓$^{\alpha}$	✓	✓	✓$^{\beta}$	n/d	n/d	n/d	n/d	✓$^{\varepsilon}$	✓	n/d
Pediatric research	✓	n/d	✓	n/d	n/d	n/d	n/d	n/d	n/d	n/d	n/d

*✓ Indicates the requirements are define in the guidelines, NR: Not required, NS: Not suitable, n/d: Not define, *except formulation, $^{\alpha}$Decision is in hands with member state, $^{\beta}$Decision is in hands with province/territory, $^{\delta}$Pharmaceutical form and formulation not define, $^{\varepsilon}$Substitution is not allowed and interchangeability is possible with practitioner's decision.*

Table 3: Comparative clinical attributes across EMA, WHO, USFDA, BGTD and BRICS-TM markets.

Clinical	EMA(EU)	WHO	USFDA (USA)	BGTD (Canada)	ANVISA (Brazil)	Russian federation (Russia)	CDSCO (India)	CFDA (China)	SAHPRA (South Africa)	TMMDA (Turkey)	COFEPRIS (Mexico)
Pharmacokinetics											
Lowest therapeutic dose	✓	✓*	✓	✓	✗	✗	✓*	✗	✓	✓	✗
Subcutaneous route	✓	✗	✓$^{\varepsilon}$	✗	✗	✗	✓	✗	✓	✓	✗
Sampling (Single dose: first & last; Multiple dose: first dose & steady state)	✓$^{\alpha}$	✓#	✓$^{\alpha}$	✗	✗	✗	✓#	✗	✓$^{\alpha}$	✓$^{\alpha}$	✗
Design (Single dose cross over; Parallel group)	✓$^{\gamma,\rho}$	✓*	✓$^{\eta\rho}$	✓	✗	✗	✓$^{\gamma,\rho}$	✓$^{\delta}$	✓$^{\gamma,\rho}$	✓$^{\gamma,\rho}$	✗
Primary parameter (Single dose: $AUC_{(0\text{-}inf)}$ Multiple dose: C_{max} & C_{trough})	✓$^{\pi}$	✗	✗	✗	✗	✗	✗	✗	✓	✓	✗
Secondary parameter (Single dose: C_{max}, T_{max}, Vss, $t_{1/2}$, Multiple dose ; $AUC_{(0\text{-}t)}$, steady state AUC)	✓	✗	✗	✗	✗	✗	✗	✗	✓$^{\mu}$	✓$^{\mu}$	✗
Acceptable range (%)	80-125	80-125	80-125	90-125$^{\omega}$	✗	✗	✓	✗	✓	✓	✗
Pharmacodynamics											
PD marker for combined PKPD	✓	✓	✓£	✓$^{\theta}$	✓	✗	✓	✓	✓	✗	✗
Fingerprinting approach (non-surrogate markers)	✓	✗	✗	Unclear	✗	✗	✗	✗	✗	✗	✗
Clinical efficacy											
Study type (Parallel, random, blinded)	✓€	✓€	✓$^{\Psi}$	✓$^{\Omega}$	✓	✗	✓	✓€	✗	✗	✗
Patient population for approved indication	✓	✓	✗	✗	✗	✗	✗	✓	✗	✗	✗
Equivalence design	✓	✓	✗	✓	✗	✗	✓	✓	✓	✗	✗
Endpoints	✓	✓	✗	✗	✗	✗	✗	✓	✗	✗	✗
Comparability margin	✓	✓	✗	✗	✗	✗	✗	✓	✗	✗	✗
Pediatric population	NR	NR	✗	✗	✗	✗	✗	✗	✗	✗	✗
Clinical safety											
Immunogenicity	✓$^{\beta}$	✓$^{\beta}$	✓	✗$^{\beta}$	✗	✗	✓	✓	✓	✗	✗
Comparative safety data	✓	✓	✓	✓	✗	✓	✓	✓	✓	✗	✗
Follow-up duration (6-12 months)	✓	✓	✗	✗	✗	✗	✗	✗	✓	✗	✗

*✓ Indicates the requirements are define in the guidelines, ✗ Indicates the requirements are not indicated in the guidelines, NR: Not required, * Higher dose for mAb clearance characteristics, $^{\alpha}$Steady state sampling for multiple doses, $^{\pi}$Additional truncated AUC for multiple doses, $^{\mu}$For multiple dose: $AUC_{(0\text{-}t)}$, steady state AUC, £ Single or Composite marker, $^{\beta}$ Double-blind, parallel analysis, duration minimum 4 weeks, €Double-blind, $^{\Omega}$Comparative trial, $^{\gamma}$Single dose for late elimination, ¥ Parallel group for mAb characteristics, $^{\rho}$Parallel group for long half-life, $^{\delta}$ Only single & multiple doses, $^{\eta}$ Cross over for short half life, $^{\varepsilon}$ Same as reference product, $^{\theta}$Study PK/PD relationship, $^{\Psi}$Only parallel design, # Single dose: Till last quantifiable concentration, $^{\omega}$equivalence margine for primary parameters.*

DISCUSSION

It is evident that mAb biosimilar exclusive guidelines are yet to be published by BRICS-TM agencies. With reference to usage of reference biological medicinal product from ICH or similarly aligned regulatory agencies, the expectations of bridging data is unclear for each of the BRICS-TM market. The necessity of proving biosimilarity post authorization, interchangeability, switching, substitution and pediatric research remains undefined with these emerging agencies. Further non-clinical and clinical mandates with detailed requirements are yet awaited. Though there are gaps in mAb biosimilar regulatory guidelines in emerging markets, we believe that the agencies are working hard to align regulatory norms in line with well-established agencies. The regulator participates in multiple forums, exchanges knowledge and is willing to upgrade. It would be advisable for the companies to approach agencies in advance to obtain biosimilar development advice so that hassle free authorization can be obtained.

ABBREVIATIONS

ANVISA: The Brazilian Health Surveillance Agency, BGTD: Biologics and Genetic Therapies Directorate, BRICS-TM: Brazil Russia India China South Africa- Turkey Mexico, CDR: Complementary Determining Region, CDSCO: Central Drugs Standard Control Organization, CFDA: China Food and Drug Administration, COFEPRIS: The Federal Commission for the Protection against Sanitary Risk, EMA: European Medicines Agency, ICH: International Conference on Harmonisation, mAb: Monoclonal Antibodies, PD: Pharmacodynamics, PK: Pharmacokinetics, RBP: Reference Biological Product, ROA: Route of Administration, SAHPRA: South African Health Products Regulatory Authority, TK: Toxico-kinetics, TMMDA: Turkish Medicines and Medical Devices Agency, USFDA: United States Food and Drug Administration, WHO: World Health Organisation

ACKNOWLEDGMENT

We highly appreciate and acknowledge 'Agnes V. Klein, MD Sr. Medical Advisor, BGTD/HC' for reviewing and supporting with expert comments for Canadian biosimilar regulations.

DISCLOSURE

Authors of this presentation have nothing to disclose.

CONTACT
HASUMATI RAHALKAR
hasumati@metinapharmconsulting.com
Tel- +91 9820113613